3-Arm RPSFT to adjust for treatment switching in superiority trials

Staburo Statistical Consulting

Chen Y[1], Esmaeili H[1], Kupas K[2], Buchner H[1]
[1]*Staburo GmbH, Aschauer Str. 26b, 81549 Munich, Germany*
[2]*Bristol-Myers Squibb GmbH & Co. KGaA, Arnulfstr. 29, 80636 Munich, Germany*

BACKGROUND

- Rapid progress in oncology and numerous approvals of medications leads to fast and frequent changes in the standard of care (SOC) during study conduct.
- In trials with more than 2 arms, both the comparison between investigational therapy (A) and former SOC (C), and the comparison between treatment (A) and alternative treatment (B), the new SOC, are of interest.
- Some subjects from arm B and arm C will cross to treatment therapy A for various reasons (eg. off-label use, existing approval of investigational drug in later line).
- Treatment switching is not only an issue for economic evaluation, but a problem for clinical assessment.
- The ITT analysis of overall survival will be biased and methods such as RPSFT need to be applied [1,2] because the underlying switching process is related to the prognosis.

METHOD

eRPSFT METHOD

- We propose an enhanced rank preserving structural failure time (eRPSFT) model to correct for treatment switching in 3-arm trials, with an adapted recensoring procedure for superiority trials (for non-inferiority trials see [2]). The eRPSFT model estimates 2 acceleration factors, for the treatment effect and for the alternative treatment effect.
- The RPSFT method is used to calculate the counterfactual survival time for each patient, where $U_{K,i}$ is counterfactual survival time and $T_{K,i}^{on}$ and $T_{K,i}^{off}$ the time with and without treatment, respectively (for patient *i* and treatment *K*).
- The acceleration factors are estimated using a grid with a range (h) from -1 to 0.5 and in increments of 0.1.
- For each value of the grid the latent survival time is calculated for every patient using the equations:

For A: $\boldsymbol{U_{(i)} = T_i^{off} + T_{A,i}^{on}(i) * exp(\alpha+\beta)}$

For B: $\boldsymbol{U_{(i)} = T_i^{off} + T_{B,i}^{on}(i) * exp(\alpha) + T_{A,i}^{On}(i) * exp(\alpha+\beta)}$

For C: $\boldsymbol{U_{(i)} = T_i^{off} + T_{A,i}^{On}(i) * exp(\alpha+\beta)}$

- The adjusted counterfactual survival time for the different treatment arms were calculated as following

For B: $\mathbf{AdjST(i)} = \boldsymbol{T_B^{On}(i) + T_A^{On}(i) * exp(\alpha+\beta) * \frac{1}{exp(\alpha)}}$

For C: $\mathbf{AdjST(i)} = \boldsymbol{T^{off}(i) + T_A^{On}(i) * exp(\alpha+\beta)}$

➢ This parameterization makes sense if A is a combination treatment of B and C (additive effect) but might need to be adapted in other settings.

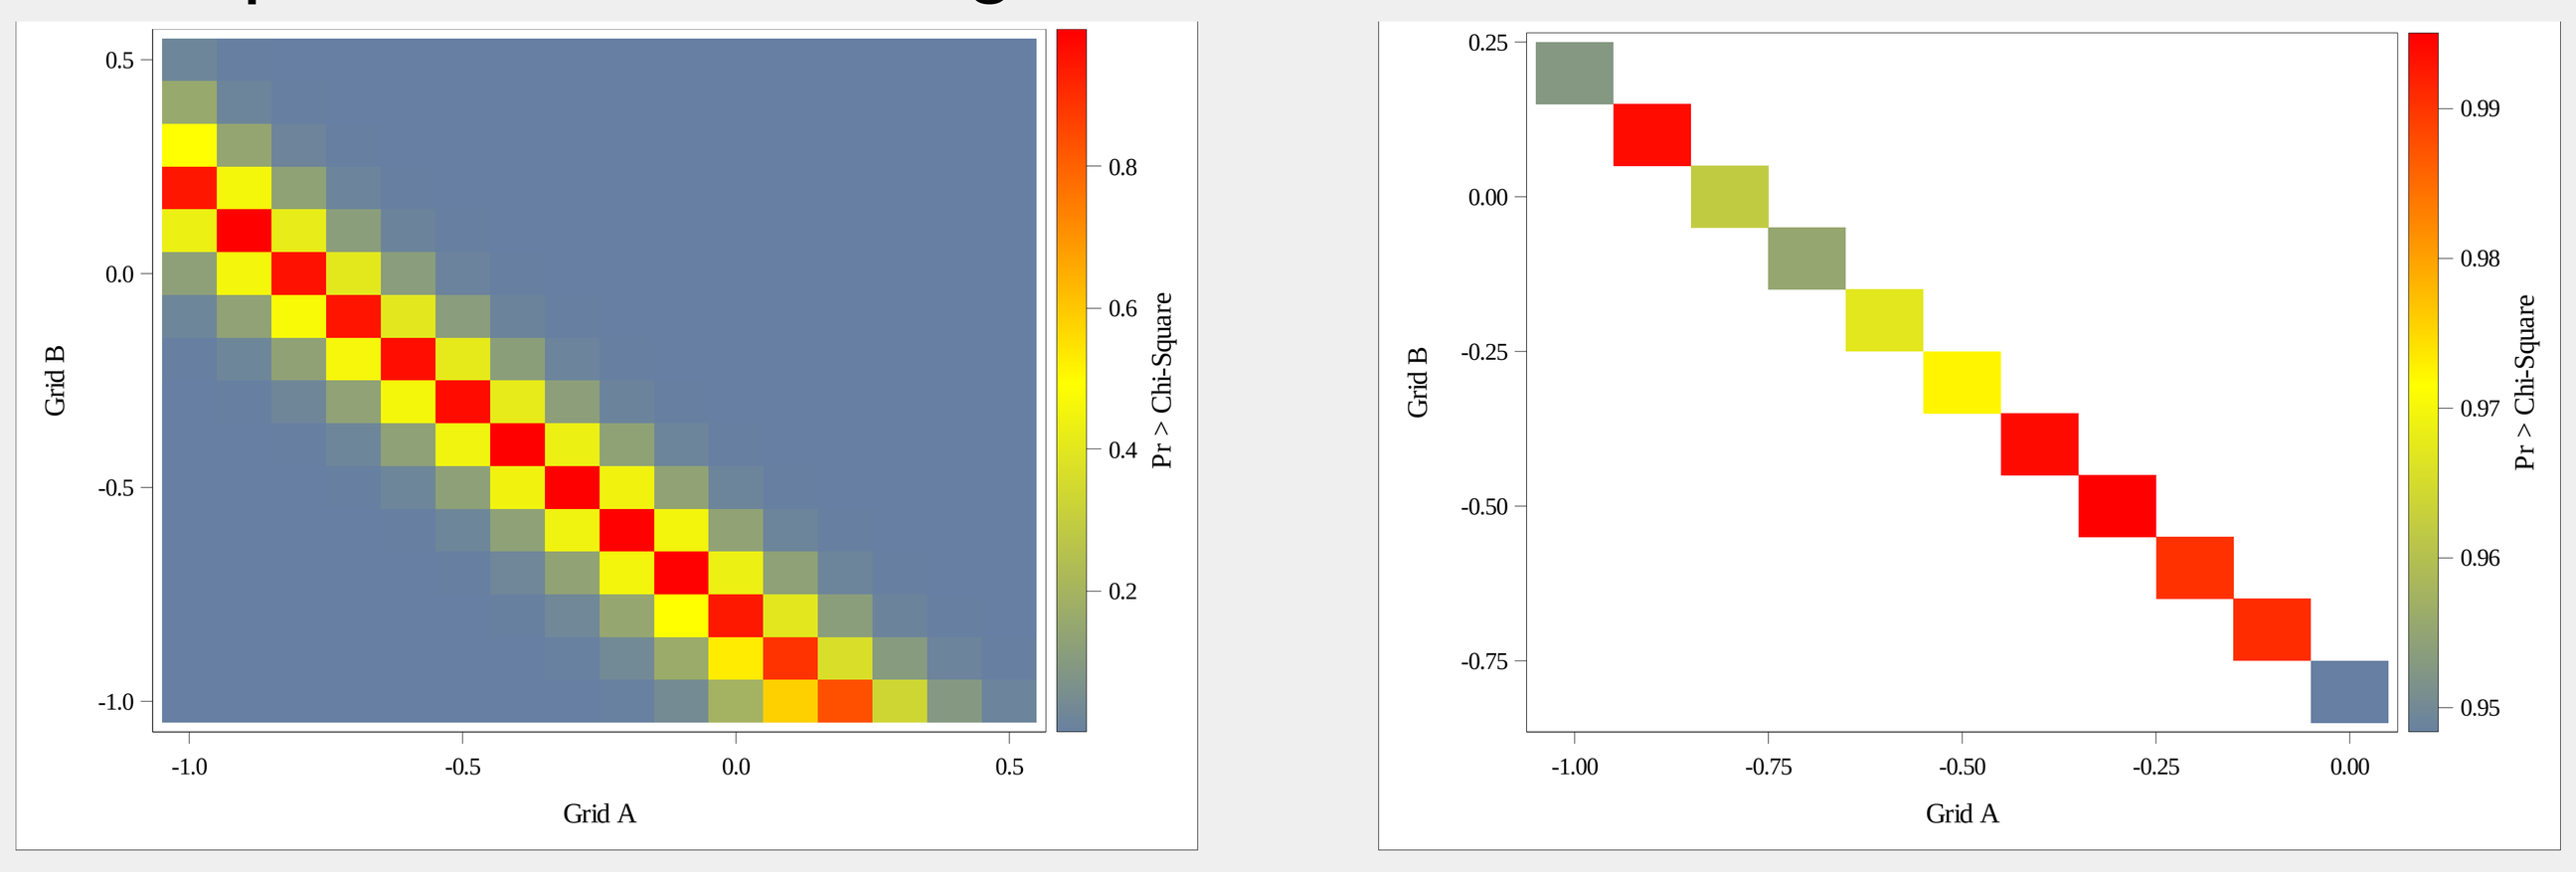
Grid A
Grid B
Pr > Chi-Square

Figure 1: *2-dimensional p-value grid for the estimation of both acceleration factors (one randomly selected simulation run). The left figure covers the whole range of values while the right figure is rescaled to visualize the maximum.*

DOUBLE-RECENSORING

- Recensoring must be performed, unless it results in significant loss of information. Not applying recensoring results in different follow-up times for the treatment arms and consequently counterfactual censoring becomes informative [3].
- First, counterfactual times are recensored by the minimum that could have been observed for each individual across their possible treatment changes during grid estimation.
- Second, in order to avoid recensoring in too many cases when treatments are beneficial, recensoring is applied again (double recensoring) to the subjects in arm B with the adjusted survival time $\boldsymbol{AdjST}$ and new censor time $\boldsymbol{C_B(i)} = \mathbf{min}[\boldsymbol{C(i),\ C(i) * \mathbf{exp}(\alpha+\beta) * \frac{1}{\mathbf{exp}(\alpha)}}]$, where $\boldsymbol{C(i)}$ is the original censor time.

SIMULATION

- A simulation study of 1000 datasets with 900 patients (300 per arm) was conducted to assess the performance of eRPSFT with 2 different mode of actions (MoA; persisting vs. treatment effect stops with last drug intake).
- The effect for A is assumed to be the best ($teffect_A = 2.0$), B is not as good as A ($teffect_B = 1.6$) but better than C. The expected true hazard ratios (HR) were 0.50, 0.625 and 0.80, respectively, for the comparisons A vs. C, B vs. C, and A vs. B.
- In scenario 1, switching rates from arm B to A are assumed to be 50% and from arm C to A 75%. In scenario 2 the rates for crossing to A are 25% for B and 50% for C.

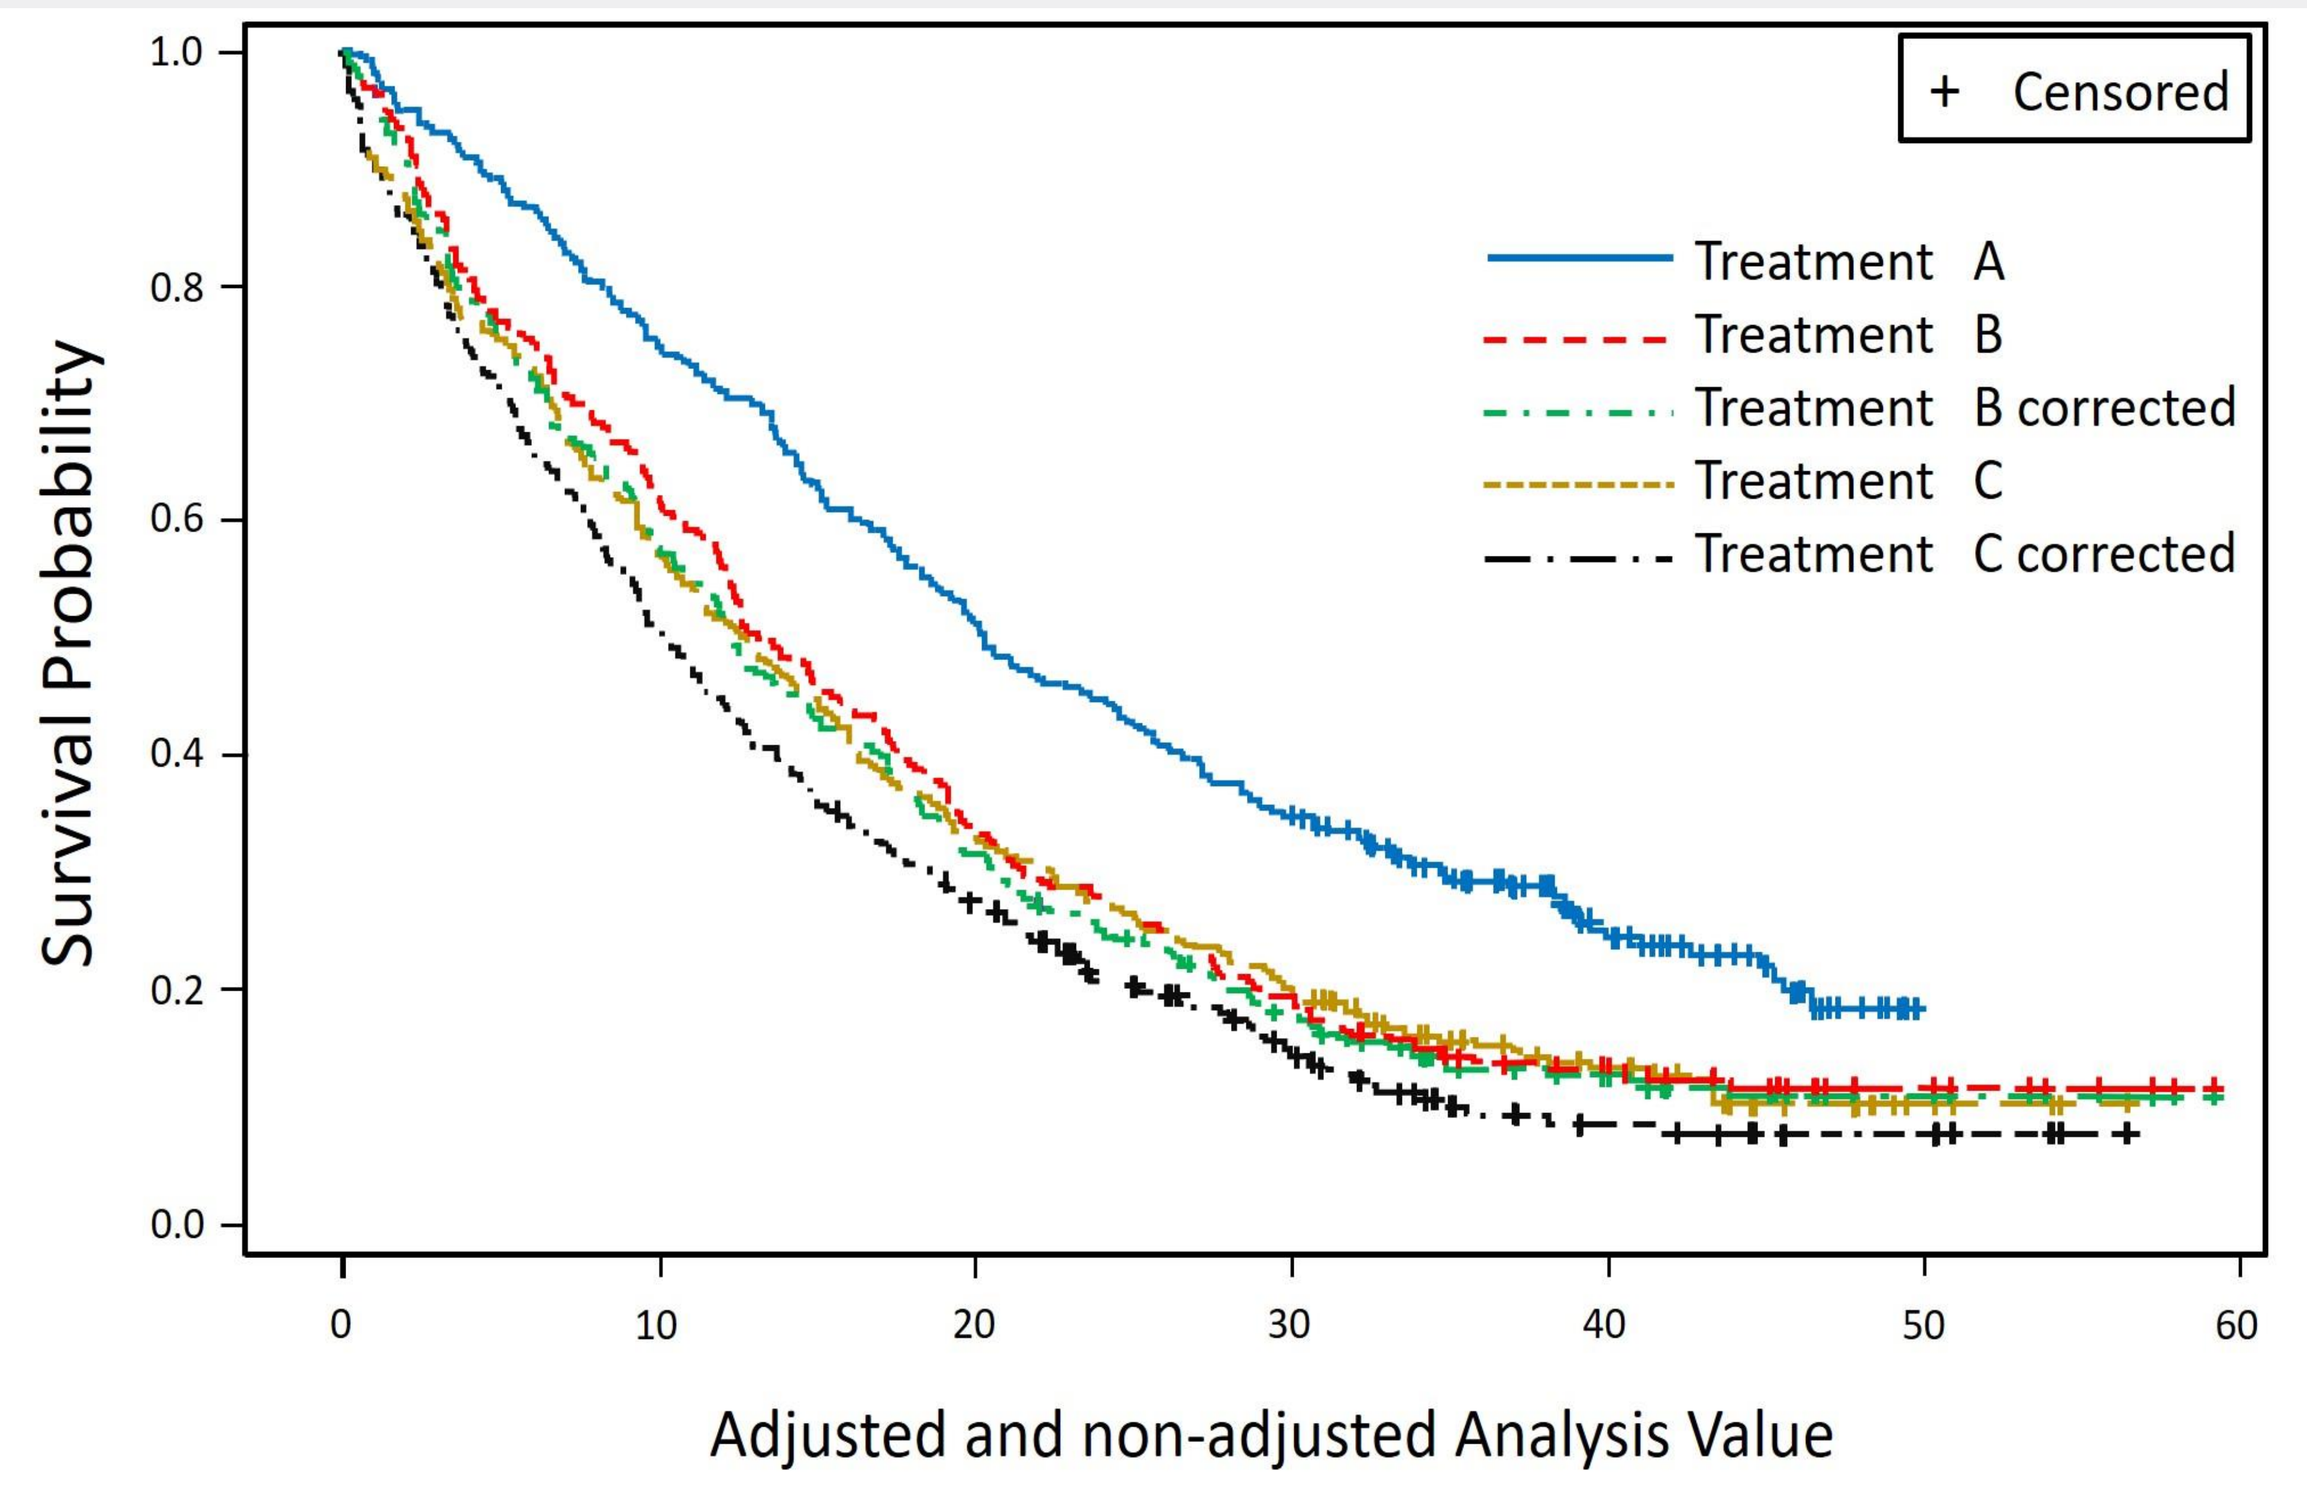
+ Censored
Treatment A
Treatment B
Treatment B corrected
Treatment C
Treatment C corrected
Survival Probability
Adjusted and non-adjusted Analysis Value

Figure 2: *The figure shows Kaplan-Meier curves for the corrected and uncorrected arms for one randomly selected simulation run of scenario 2 with treatment effect stops.*

RESULTS

- The eRPSFT HRs for A vs. C were 0.512 (2.5th and 97.5th percentiles [CI]: 0.419, 0.627) and 0.507 (0.414, 0.621) in the scenarios, where the treatment effect persists. The estimates for the scenarios with treatment effect stops were 0.569 (0.467, 0.694), and 0.540 (0.443, 0.659).
- The HRs for B vs. C were 0.604 (0.453, 0.845) and 0.614 (0.504, 0.749) in the scenarios where the treatment effect persists, and 0.710 (0.584, 0.863) and 0.610 (0.500, 0.743) where the treatment effect stops.
- The HRs for A vs. B were 0.859 (0.608, 1.123) and 0.830 (0.686, 1.004) in the scenarios where the treatment effect persists, and 0.807 (0.668, 0.975) and 0.905 (0.750, 1.093) where the treatment effect stops.

Table 1: *Results of both simulation scenarios. Comparing eRPSFT results with true values and naïve ITT results.*

	eRPSFT HR (95% CI)	Bias	True HR	ITT HR (95% CI)
Scenario 1 - treatment effect persists				
A vs C	0.512 (0.419, 0.627)	0.012368	0.5	0.684 (0.579, 0.819)
B vs C	0.604 (0.495, 0.736)	0.021354	0.625	0.802 (0.669, 0.955)
A vs B	0.859 (0.710, 1.039)	0.058981	0.8	0.857 (0.709, 1.027)
Scenario 1 - treatment effect stops				
A vs C	0.569 (0.467, 0.694)	0.069333	0.5	0.749 (0.626, 0.898)
B vs C	0.710 (0.584, 0.863)	0.084768	0.625	0.833 (0.698, 0.898)
A vs B	0.807 (0.668, 0.975)	0.006804	0.8	0.903 (0.744, 1.086)
Scenario 2 - treatment effect persists				
A vs C	0.507 (0.414, 0.621)	0.006812	0.5	0.619 (0.520, 0.724)
B vs C	0.614 (0.504, 0.749)	-0.01075	0.625	0.751 (0.633, 0.894)
A vs B	0.830 (0.686, 1.004)	0.029829	0.8	0.828 (0.688, 0.986)
Scenario 2 - treatment effect stops				
A vs C	0.540 (0.443, 0.659)	0.040044	0.5	0.619 (0.518, 0.733)
B vs C	0.610 (0.500, 0.743)	-0.10508	0.625	0.753 (0.624, 0.891)
A vs B	0.905 (0.750, 1.093)	0.105181	0.8	0.826 (0.690, 0.979)

Bias is the difference between eRPSFT estimation and true HR.

CONCLUSIONS

- The eRPSFT performed well with small bias for all calculations.
- All eRPSFT CIs contain the true HR; the precision of RPSFT increases with decreasing switching rate; MoA should be considered since it shows an (ambiguous) influence on the results.
- The uncorrected HRs under-estimate the true effect in most situations and show similar performance (especially bias) as eRPSFT only in very few comparisons.
- Only few ITT CIs contain the true HR (only A vs. B, all other CI exceed the true HR).
- The eRPSFT improves the estimation compared to a naïve ITT analysis and should be used to adjust for treatment switching in 3-arm superiority trials.

ACKNOWLEDGEMENTS

The work was supported financially by Bristol-Myers Squibb.

REFERENCES

[1] Robins JM, Tsiatis AA. Correcting for non-compliance in randomized trials using rank preserving structural failure time models. *Communications in Statistics* 1991; 20:2609–2631.
[2] Ying Wu, Liang Zhao, Yan Hou, Kang Lia and Xiaohua Zhou: Correcting for non-compliance in randomized non-inferiority trials with active and placebo control using structural models, Statistics in Medicine 2015, 34:950–965
[3] White I.R. Letter to the editor: Survival analysis of randomized clinical trials adjusted for patients who switch treatments by M. G. Law and J. M. Kaldor, Statistics in Medicine, 15, 2069–2076 (1996)

Staburo GmbH, Aschauer Str. 26b, 81549 Munich, info@staburo.de, https://www.staburo.de